Emerging From Burnout: Keeping PACU Nurses Engaged After COVID

Wendy Ferguson, BSN, RN, CEN

Background

Perioperative nurses continue to face mental and physical stress and exhaustion in the workplace post-COVID. Nurses have continued to work with staff shortages, greater patient loads, supply shortages, financial recovery restrictions all of which have exacerbating preexisting crisis levels of burnout, and low sense of personal accomplishment among the nursing profession (ANA, 2024; Pouge et. all, 2022; CDC, 2023). An estimated 52% of registered nurses surveyed in 2022 plan to leave their nursing position due to workplace stressors (ANF, 2022). Nurses in the perioperative setting at Kootenai Health have worked with fellow staff members and nursing leadership to create an environment that emphasizes self-care, work-life balance and control of schedule to increase nursing moral and staff retention.

Setting

Located in northern Idaho, Kootenai Health is a 397-licensed bed, non-profit community hospital additionally supporting six rural access hospitals in the area. Kootenai Health is a Verified Level II ACS trauma center, with 55,984 ER visits, 2067 traumas and 9340 surgeries completed in 2023. The surgical department consist of 13 OR suites, 17 PACU bays, 24 Pre-Op bays, and five Extended Recovery Unit (ERU) bays. Kootenai Health is also a recognized Magnet hospital, Level III NICU center, and Gallup Great Workplace award recipient (Kootenai Health, 2024).

Project Objective

To decrease perioperative nurse burnout and increase nurse retention by identifying stressors of greatest concern, then implementing strategies that increase awareness, education, and overall moral in nurses in the perioperative setting.

Process of Implementation

From spring 2022 to the fall of 2023, the perioperative unit clinical coordinators, UPC members and unit manager discussed perioperative nurse burnout factors and monthly perioperative staff meetings, unit staffing committee meetings and Unit Practice Council (UPC) meetings. Discussions included the perioperative nurse's concern to maintain control of their work schedule through a self-scheduling method, that did not involve in-person sign up. In the fall of 2023, the PACU staffing committee and department coordinators distributed surveys in order to exclusively move to an online Google Docs scheduling platform to accommodate self-scheduling concerns.

Statement of Successful Practice

With the identification and recognition of stressors resulting in burnout within their unit, perioperative staff nurses worked with department coordinators and managers to create ways to decrease burnout and increase staff satisfaction and retention. Staff surveys were distributed via email and written paper to help identify the greatest stressors and direction for change. After a review of anonymous department survey results regarding schedule preferences and individual stressors, the UPC trialed new practices include the implementation of an online self-schedule or block schedule template; cross-training of perioperative nurses in Pre-Op, PACU Phase I, Phase II and Extended Recovery Unit (ERU) areas; optional nursing points-based clinical ladder program for nurse recognition with financial rewards; hospital-wide points-based wellness program; and encouraged use of scheduled paid time off (PTO) for self-care and wellness. Available hospital resources for stress reduction and staff retention were also identified and distributed to staff via emails, unit monthly meetings, and posted break room flyers.

Survey of Staff and Findings:

- Between 2022 and 2024, electronic and printed surveys were distributed to 62 perioperative staff RNs regarding block or self-schedule preferences. An estimated 50% of unit staff RNs responded to the pre-schedule change surveys.
- Six months after the new on-line, self-scheduling process was implemented, a follow-up survey was emailed to unit staff for feedback. The survey forms were completed anonymously by staff and placed in a designated folder to ensure privacy. 82% of the unit RNs responded.
- Using a rating scale where 1 = dissatisfied, 5 for neutral, 10 = very satisfied, and n/a does not apply, staff members responded an average satisfaction score of 9 regarding preference for block schedule, self-scheduling and on-line scheduling options. A rating of 8 was reported regarding choice of own on-call schedule, 6 for assigned on-call and 3 for in-person schedule sign ups.
- 97% percent of staff responded "Yes" to the question, "Do you feel that having choices in your schedule has improved your job satisfaction?"
- The results were tallied using a simple quantitative method and presented to department manager, clinical coordinators, and staffing committee for review.
- Available hospital well-being resources were identified, re-visited, and distributed to perioperative staff including free hospital counseling services, Vital WorkLife peer coaching, Kootenai Health Family Fund, Schwartz Rounds, Unmind App, and MyHealth wellness initiatives including financial reimbursement.
- Perioperative nurses were also encouraged to attend optional in-house groups, such as hospital trauma focus groups. These groups were established to encourage front line nurses to have a voice and express concerns in their work environment.

Figure 1: Schedule Survey Results

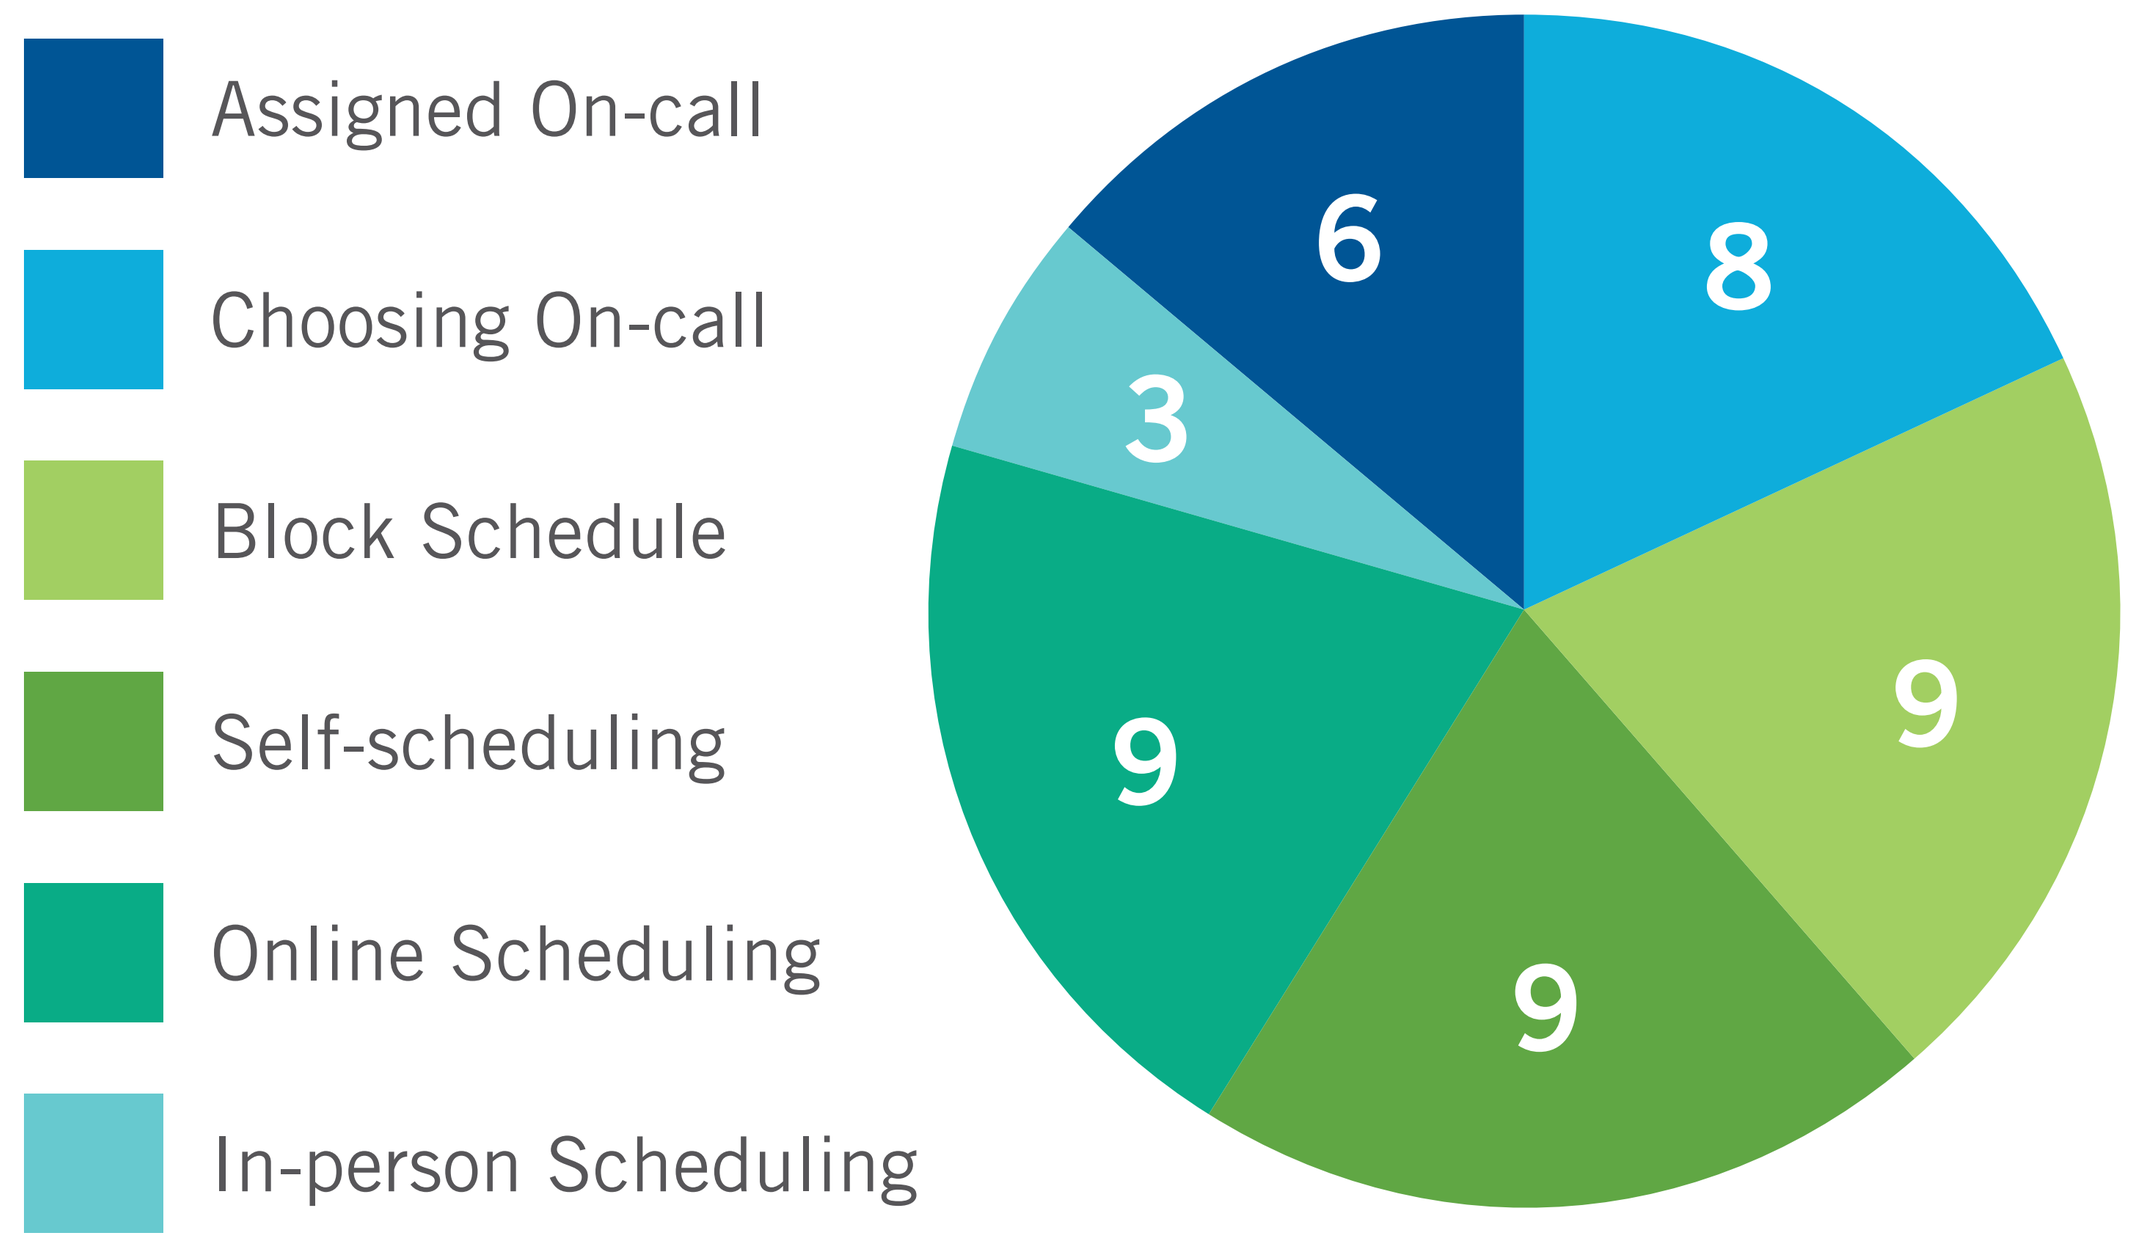

Assigned On-call
Choosing On-call
Block Schedule
Self-scheduling
Online Scheduling
In-person Scheduling
6
8
3
9
9
9

Implications for Advancing the Practice of PeriAnesthesia Nursing

Avoiding nursing burnout and retaining staff begins with awareness. Perioperative nurses in this unit self-reported an overall increase in personal moral and job satisfaction by having more control of their work-life balance though self-scheduling, optional clinical ladder programs, scheduled use of PTO, intradepartmental cross-training, hospital-wide wellness resources and an open-door management environment. Perioperative units will likely continue to face staffing shortages and factors of nurse burnout. However, when perioperative unit leaders and nurses work together to identify work stressors, implement self-scheduling programs, and revisit available hospital resources, a positive environment of trust and healing begins that can directly aid in decreasing perioperative burnout and increasing staff retention.

Successful Practice Strategies:

- **Schedule:** online self-scheduling, staff surveys for preferences, clear communication of on-call expectations
- **Pay:** financial incentive rewards programs including clinical ladder (Nurse Excellence) and health wellness (MyHealth), certification pay, and education assistance
- **Self-care:** scheduled time off encouraged, work-life balance, hospital wellness resources
- **Staffing:** balanced staffing ratios, cross training in other perioperative areas, open-door of communication with manager and unit coordinators

Figure 2: Key Points

- Online Self-scheduling
- Financial Incentive Programs
- Balanced Staffing Ratios
- Clear Communication
- Self-care and Scheduled Time Off

References

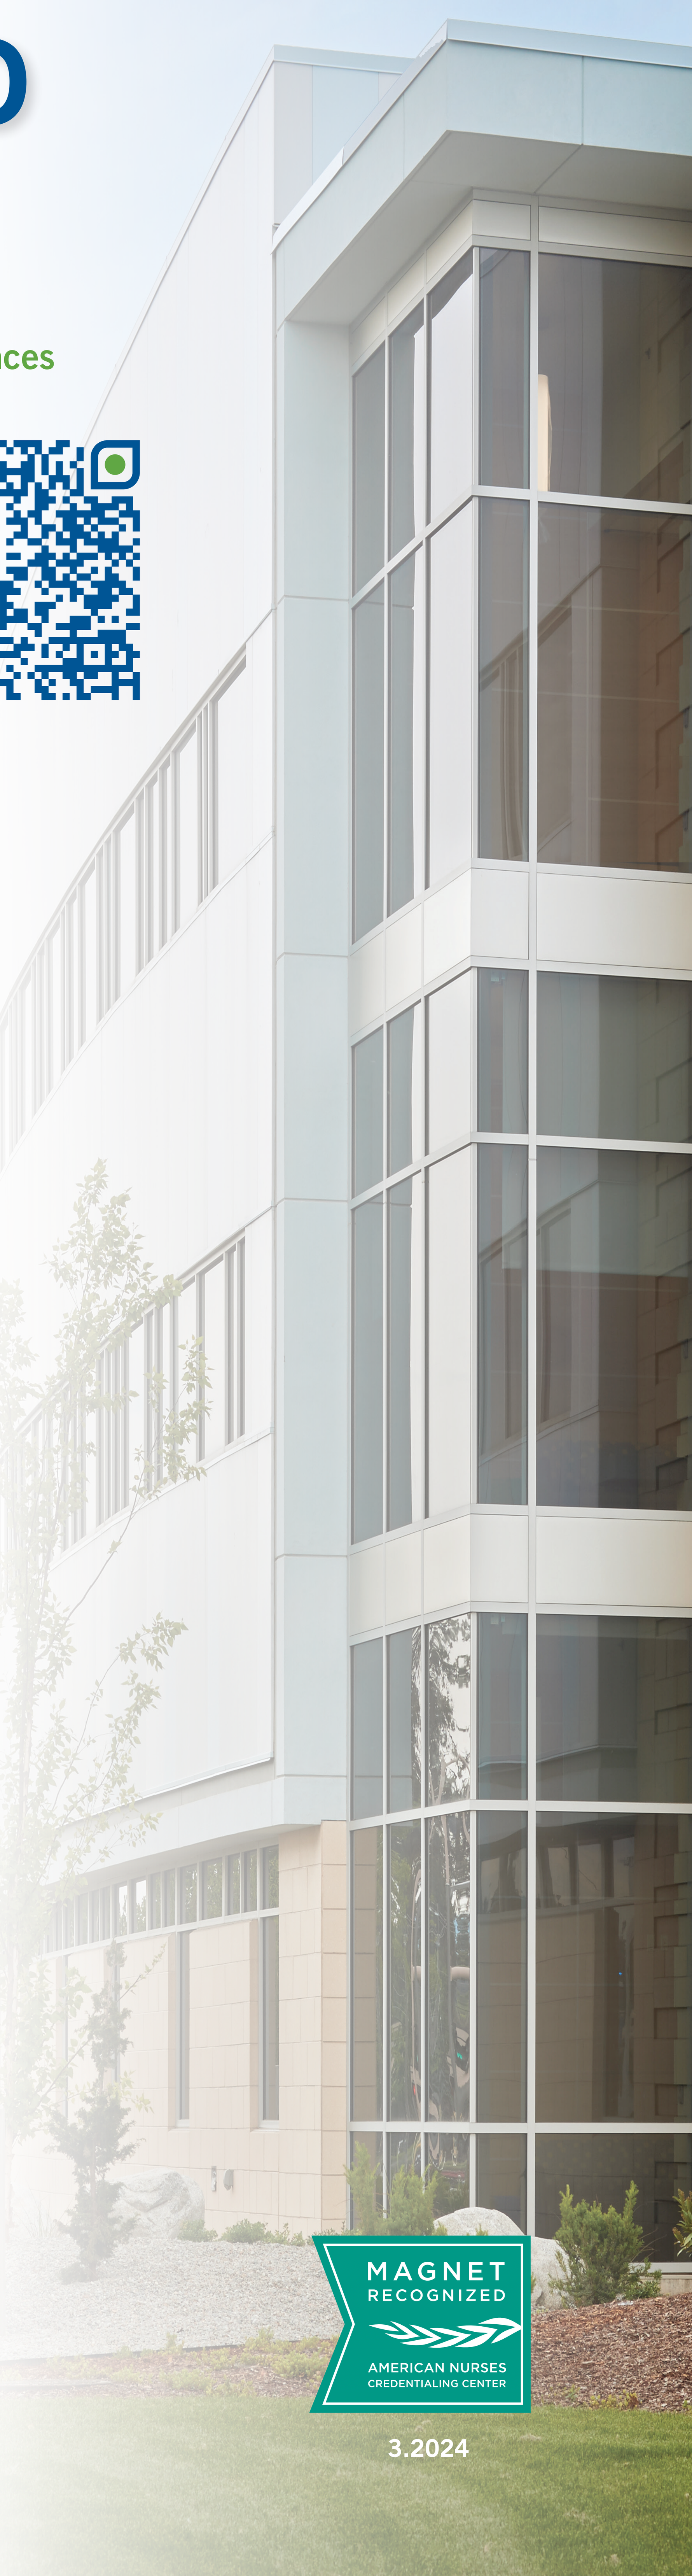

MAGNET RECOGNIZED
AMERICAN NURSES CREDENTIALING CENTER
3.2024

KootenaiHealth

kh.org/nursing | 208.625.4000

2003 Kootenai Health Way
Coeur d'Alene, ID 83814